The Cardiolipin-Targeting Compound SBT-272 Attenuates Neurodegeneration, Delays the Onset of Neurological Signs and Extends Lifespan in Male, SOD1 G93A Transgenic Mice

Dennis Keefe[1], Guozhu Zheng[1], Inese Smukste[1], Arie Mobley[2], Laurent Bogdanik[2], Mark Bamberger[1]

[1]Stealth Biotherapeutics, Newton, MA; [2]Jackson Laboratories, Bar Harbor, Maine

INTRODUCTION

- Mitochondrial dysfunction is believed to play a prominent role in the pathophysiology of Amyotrophic Lateral Sclerosis (ALS)
- SBT-272 is an investigative molecule currently in preclinical development for the treatment of patients with rare neurodegenerative disease through the promotion of mitochondrial health
 - SBT-272 preserves the inner mitochondrial membrane structure by interacting with the phospholipid cardiolipin, a critical mediator of the mitochondrial cristae network, and key structural and functional element of mitochondrial respiratory super-complexes
- Through its protective effect on cardiolipin, SBT-272 has been shown to reduce the generation of harmful reactive oxygen species (ROS) while also increasing the efficiency of electron transport under conditions of stress, leading to a net gain in bioenergetic output and a decrease in oxidative damage
 - Importantly, both mechanisms - damage from ROS and bioenergetic deficit - have been described in the mitochondria obtained from subjects with ALS

OBJECTIVE

- To investigate the potential efficacy of SBT-272 in treating ALS, mice expressing the mutant human SOD1 G93A transgene were treated with daily intraperitoneal injections of SBT-272 starting at 8 weeks of age
 - Consistent with the onset of neuromuscular dysfunction in mice as measured through electromyography

METHODS

- A total of 108 mutant human transgenic SOD1 mice (B6SJL-Tg(SOD1*G93A)1Gur/J) were used for the study
- Animals were assigned to one of two groups, one dedicated to histology and one dedicated to behavior analyses and lifespan
 - Dosing began when animals reached 8 weeks of age
- Animals in the dedicated histology group were treated with daily intraperitoneal injections of drug or vehicle for six weeks as follows: vehicle (6 male/6 female); SBT-272 at 0.5mg/kg (6 male/6 female); SBT-272 at 5.0mg/kg (6 male/6 female)
 - Histological analyses are pending and will include sciatic nerve axon counts, spinal cord Iba-1 and GFAP-1 staining, spinal cord neuromuscular junction occupancy, and electron microscopy of spinal cord sections to investigate mitochondrial morphology
- Animals in the dedicated behavioral/lifespan group were treated with daily intraperitoneal injections of drug or vehicle until humane end of life as follows: vehicle (10 male/10 female); SBT-272 at 0.5mg/kg (10 male/10 female); SBT-272 at 5.0mg/kg (10 male/10 female)
- In-life observations included daily weight gain, weekly forelimb grip strength, and weekly neurological scoring, which was assessed using a four-point qualitative ratings scale
- Bi-weekly retro-orbital blood draws were performed to verify drug exposure and for biomarker analyses

RESULTS

- Treatment of symptomatic ALS mice resulted in therapeutic benefit that was sex dependent
- Male mice treated with high dose of drug experienced a dose-dependent delay in the onset of neurological symptoms, as assessed by a qualitative ratings scale, and a dose-dependent survival benefit that was highly correlative with plasma neurofilament levels (extended lifespan) (**Figure 1**)
 - Both results were statistically significant
- Decline in grip strength was attenuated in male mice treated with high dose of drug, although these results were not statistically significant (**Figure 2**)
- Circulating plasma levels of the neuroinflammatory biomarker neurofilament light chain, an emerging biomarker of axonal damage, were significantly decreased at high dose of drug – these results were statistically significant (**Figure 3**)
- Notably, survival and biomarker levels were highly correlative in male mice (**Figure 4**)
 - Female SOD1 G93A mice, which present with a milder phenotype and displayed lower levels of the circulating neurofilament biomarker, were not affected by drug despite comparable levels of exposure
- Confirmatory histology studies are on-going and are expected to corroborate the in-life and biomarker observations

Figure 1. Effect on Neurological Symptoms and Survival

SBT-272 at a dose of 5.0mg/kg delayed the progression of neurological symptom onset in male animals relative to vehicle treated animals, as determined by two-way ANOVA (**A**). In addition, the lifespan of male animals dosed with SBT-272 at 5.0mg/kg was significantly increased compared to vehicle control (**B**). There were no effects in female mice, which present with a milder disease phenotype in this transgenic model.

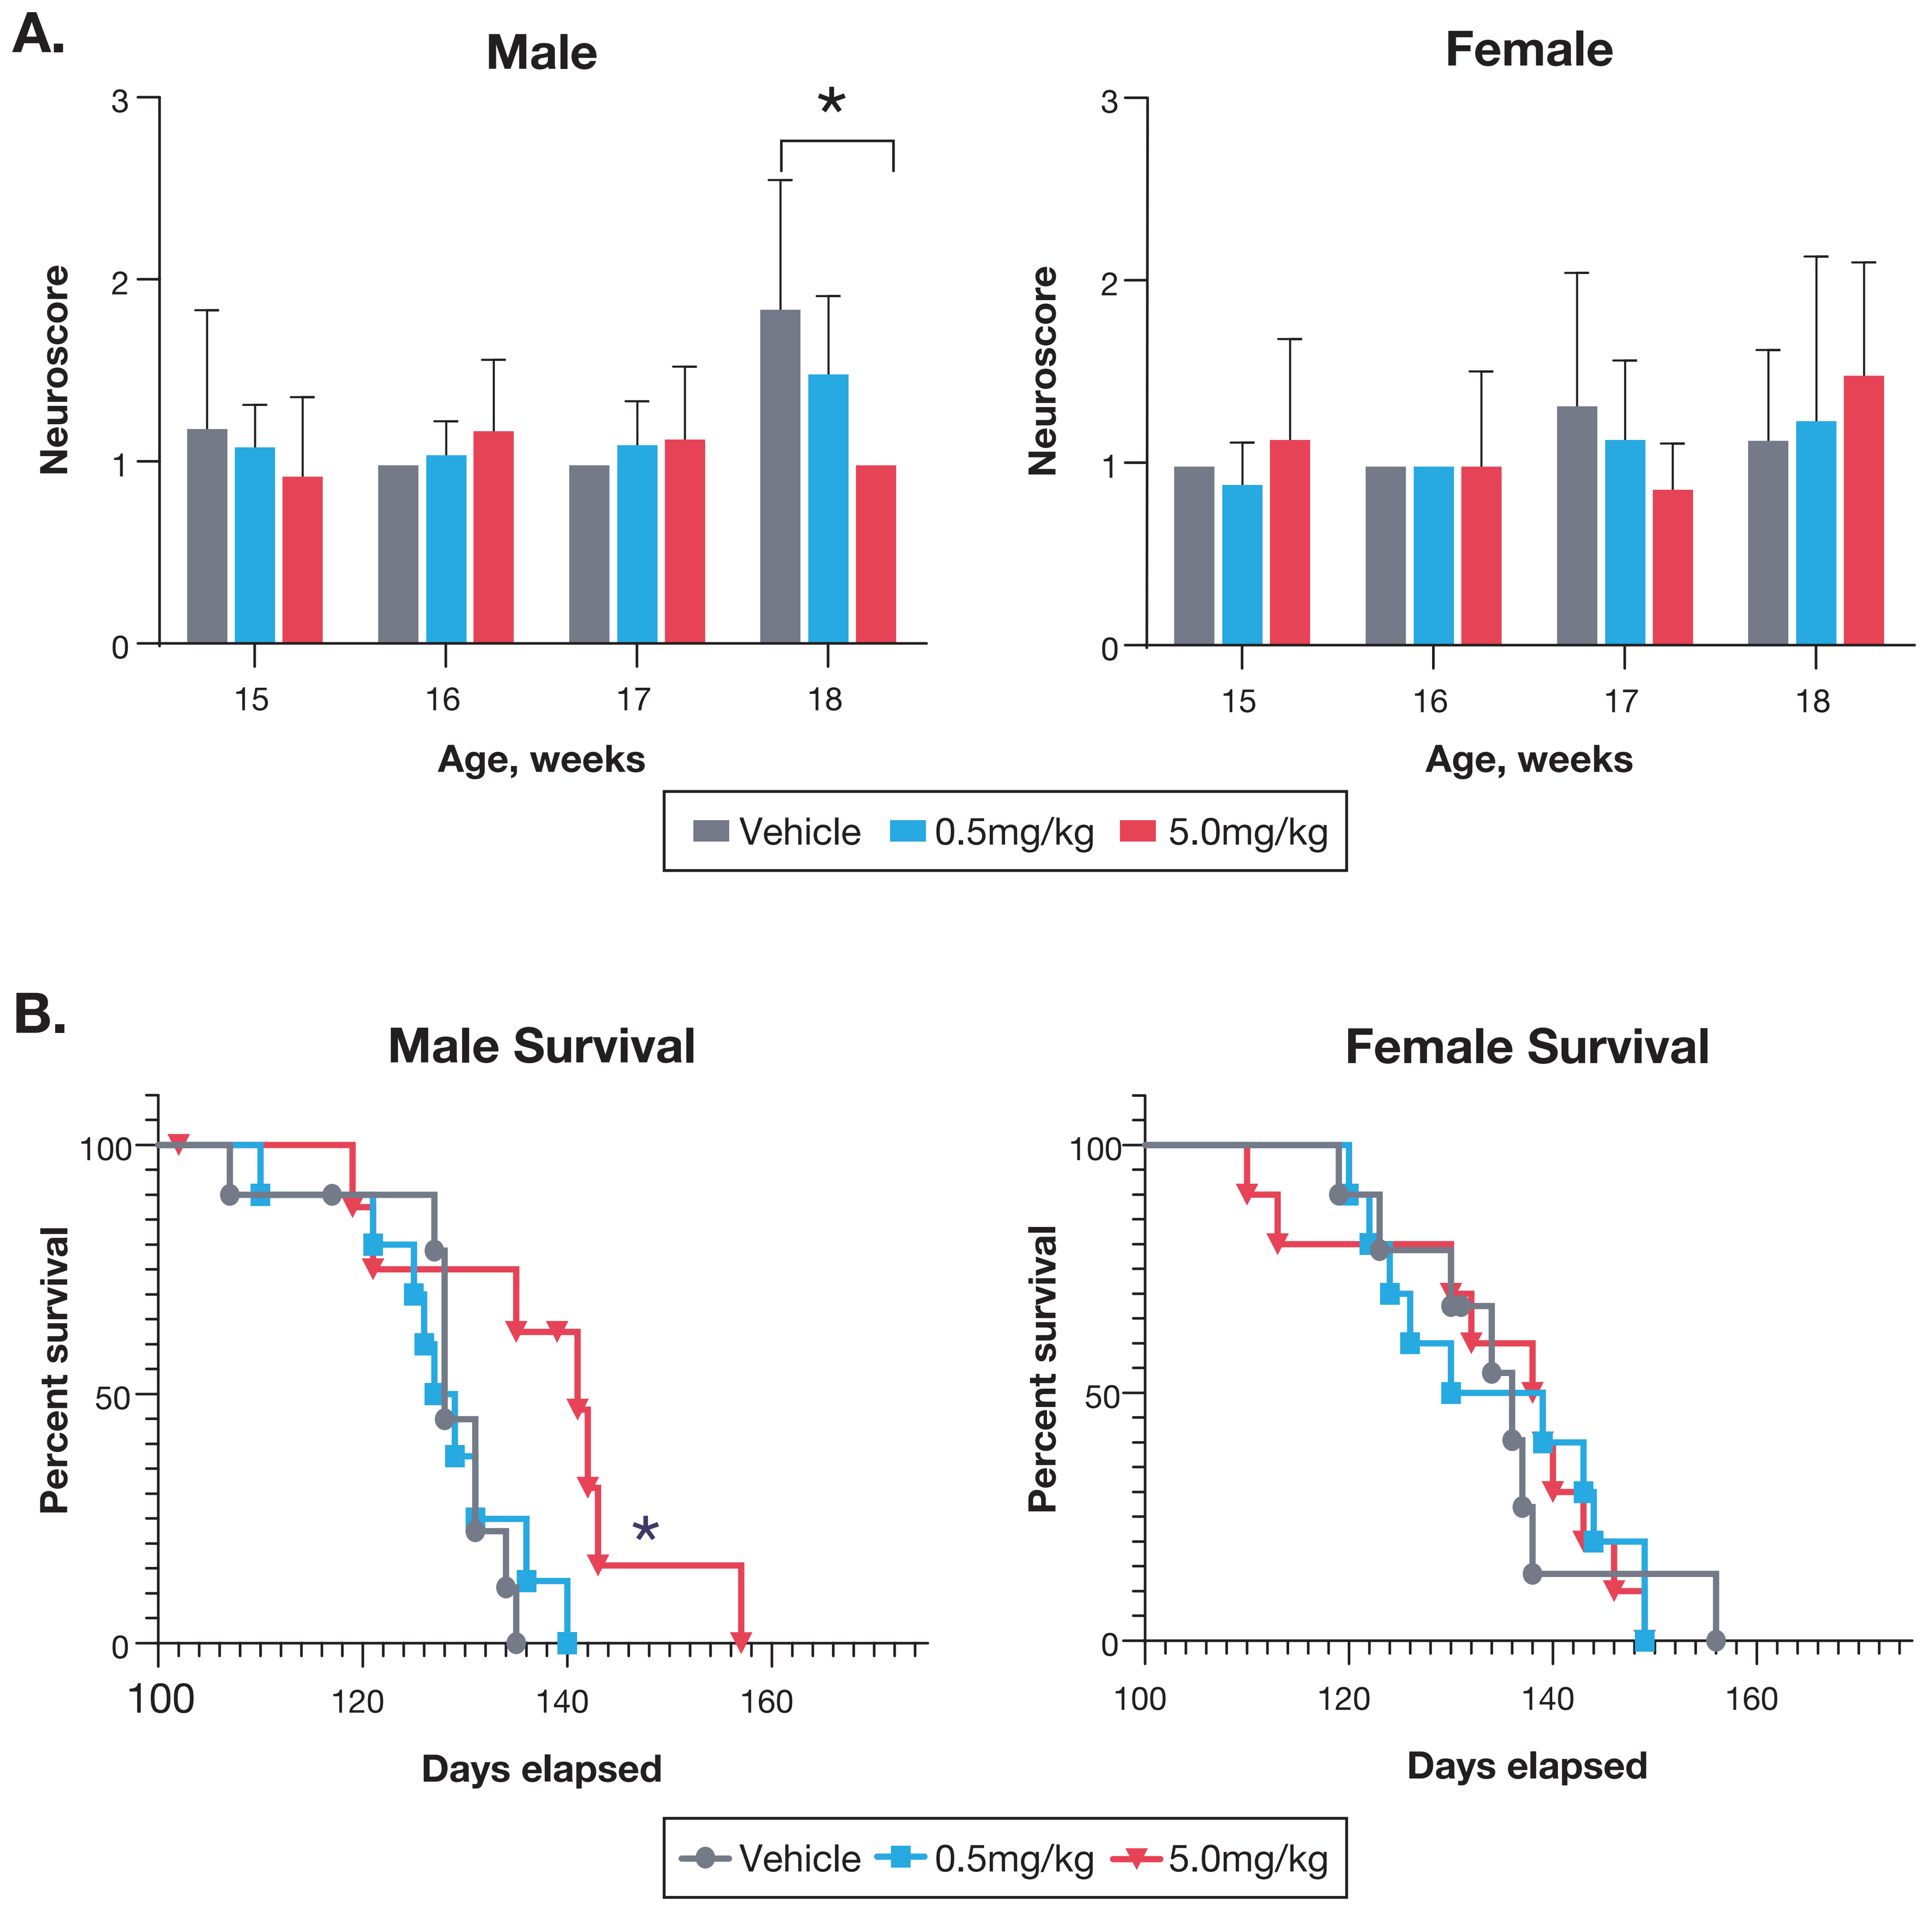
A.
Male
Female
Neuroscore
Age, weeks
Vehicle
0.5mg/kg
5.0mg/kg
B.
Male Survival
Female Survival
Percent survival
Days elapsed

Figure 2. Effect on Grip Strength

The mean of individual animal decline is depicted for animals following 10 weeks on drug. After this time the group sizes became unbalanced due to animal loss and data become difficult to interpret. Male animals treated with high dose (5.0mg/kg) SBT-272 demonstrated a trend in protection from loss of grip strength (**A**), although the magnitude of effect falls just short of statistical significance (p=0.08). No such trend was apparent in female mice (**B**).

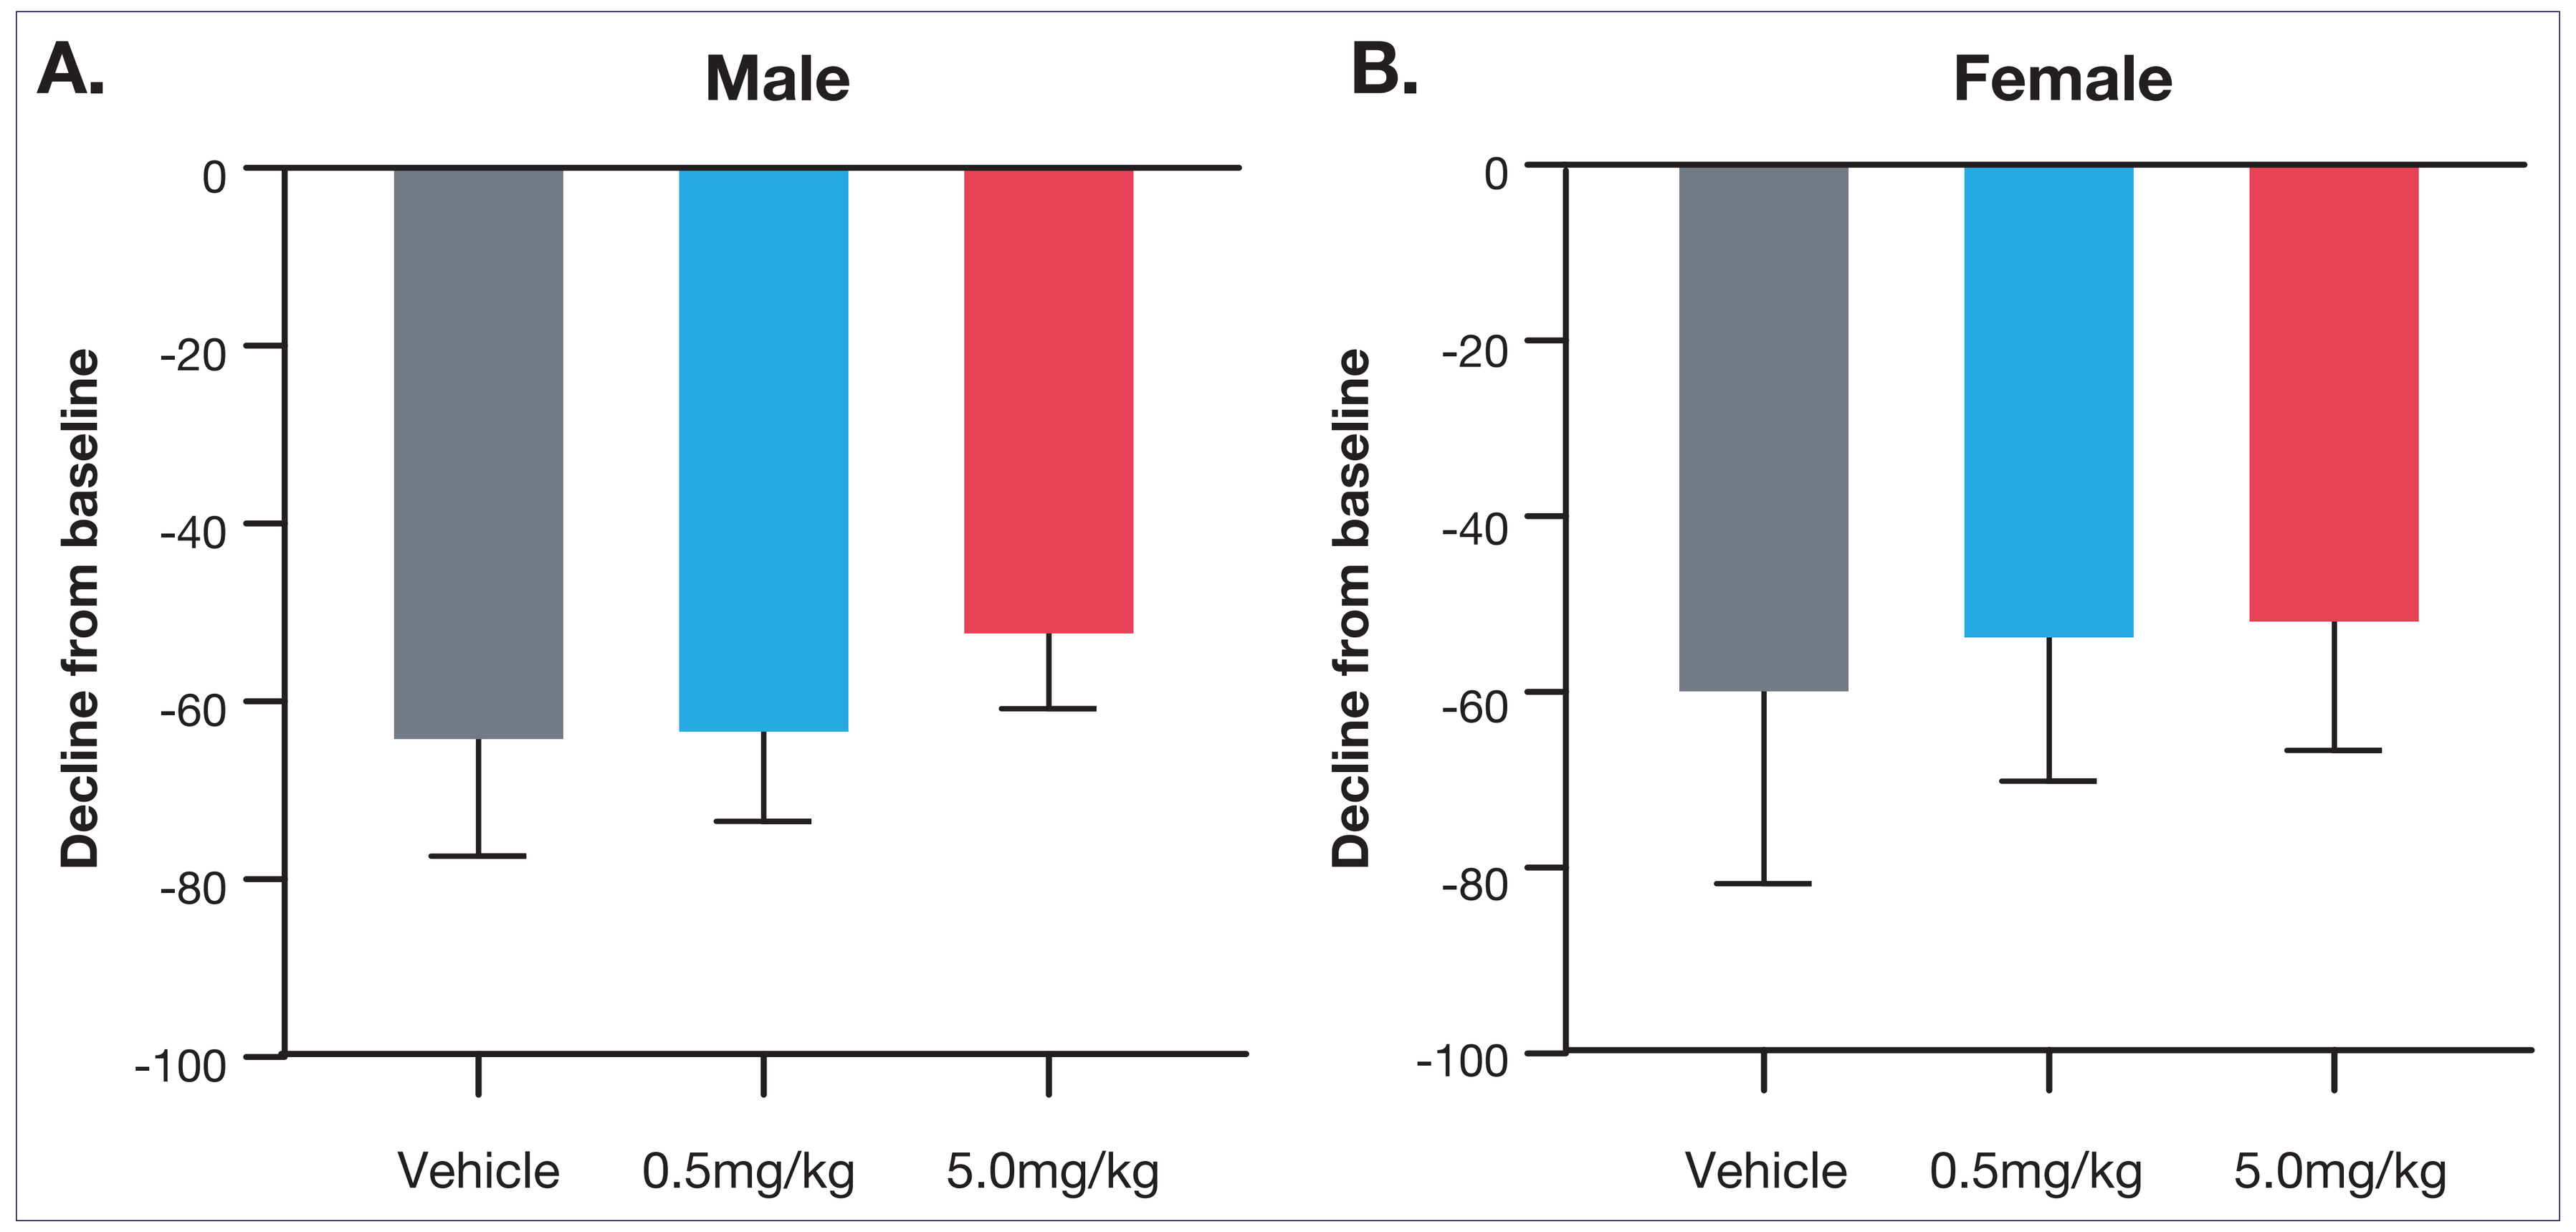
A.
Male
B.
Female
Decline from baseline
Vehicle
0.5mg/kg
5.0mg/kg

Figure 3. Effect on Plasma Levels of Neurofilament Light Chain

Plasma levels of Neurofilament Light Chain (NfL), a marker of axonal damage, following 10 weeks of SBT-272 or vehicle control administration. SBT-272 at 5.0mg/kg had a statistically significant impact on the accumulation of NfL in male transgenic mice, as determined by one way ANOVA, suggesting protection from axonal damage in the CNS (**A**). Note that female mice had lower overall levels of NfL, consistent with the milder disease phenotype associated with female mice in this model (**B**).

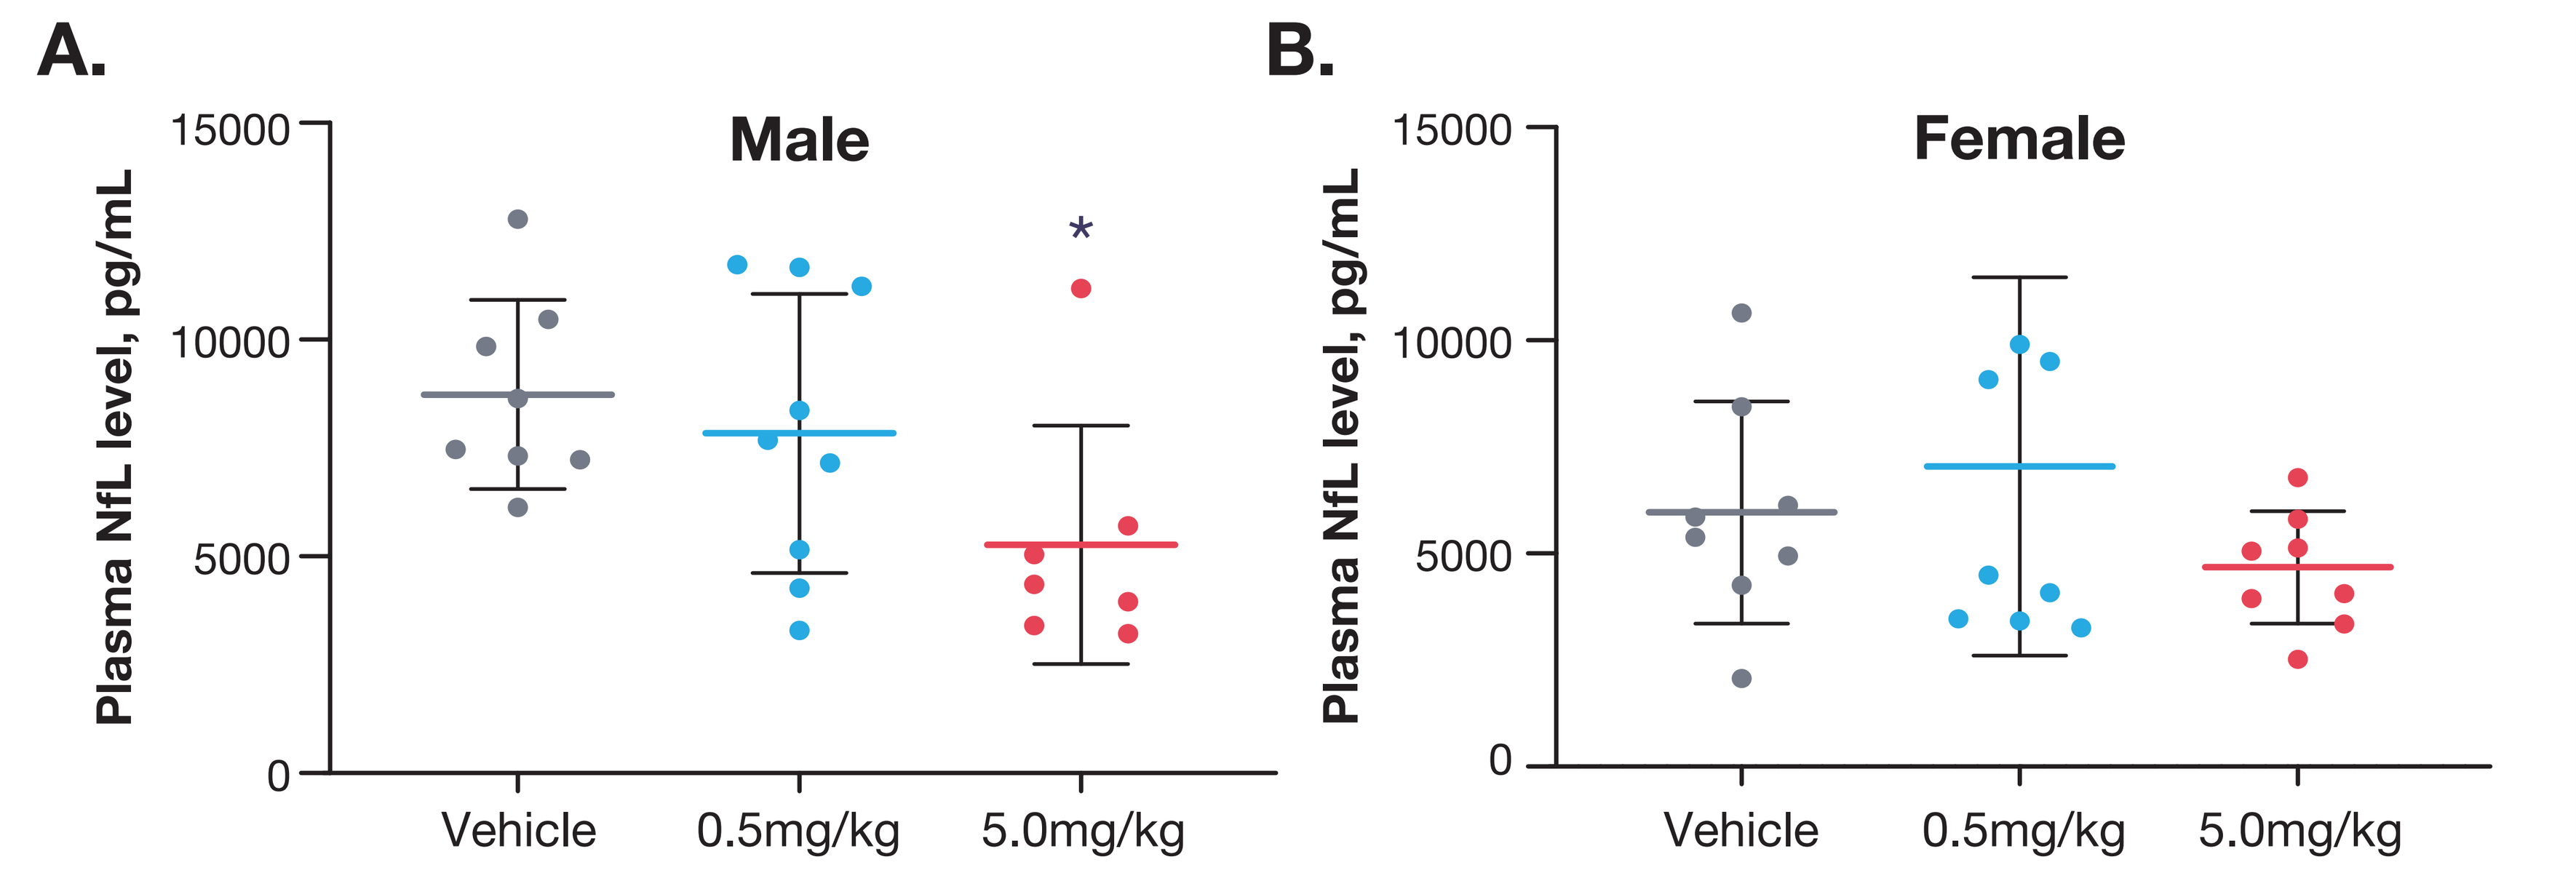
A.
Male
B.
Female
Plasma NfL level, pg/mL
Vehicle
0.5mg/kg
5.0mg/kg

Figure 4. Correlation Between Plasma Neurofilament Levels and Survival

There is a significant correlation between plasma neurofilament levels and animal survival in male SOD1 G93A transgenic mice. Depicted are plasma NfL levels for every male mouse in this study plotted as a function of their age at humane end of life. The correlation between accumulation of the axonal damage biomarker and animal lifespan is highly significant, suggesting that SBT-272 prolongs the lifespan of SOD1 G93A mice at least partly through protection of axonal loss or damage in the CNS.

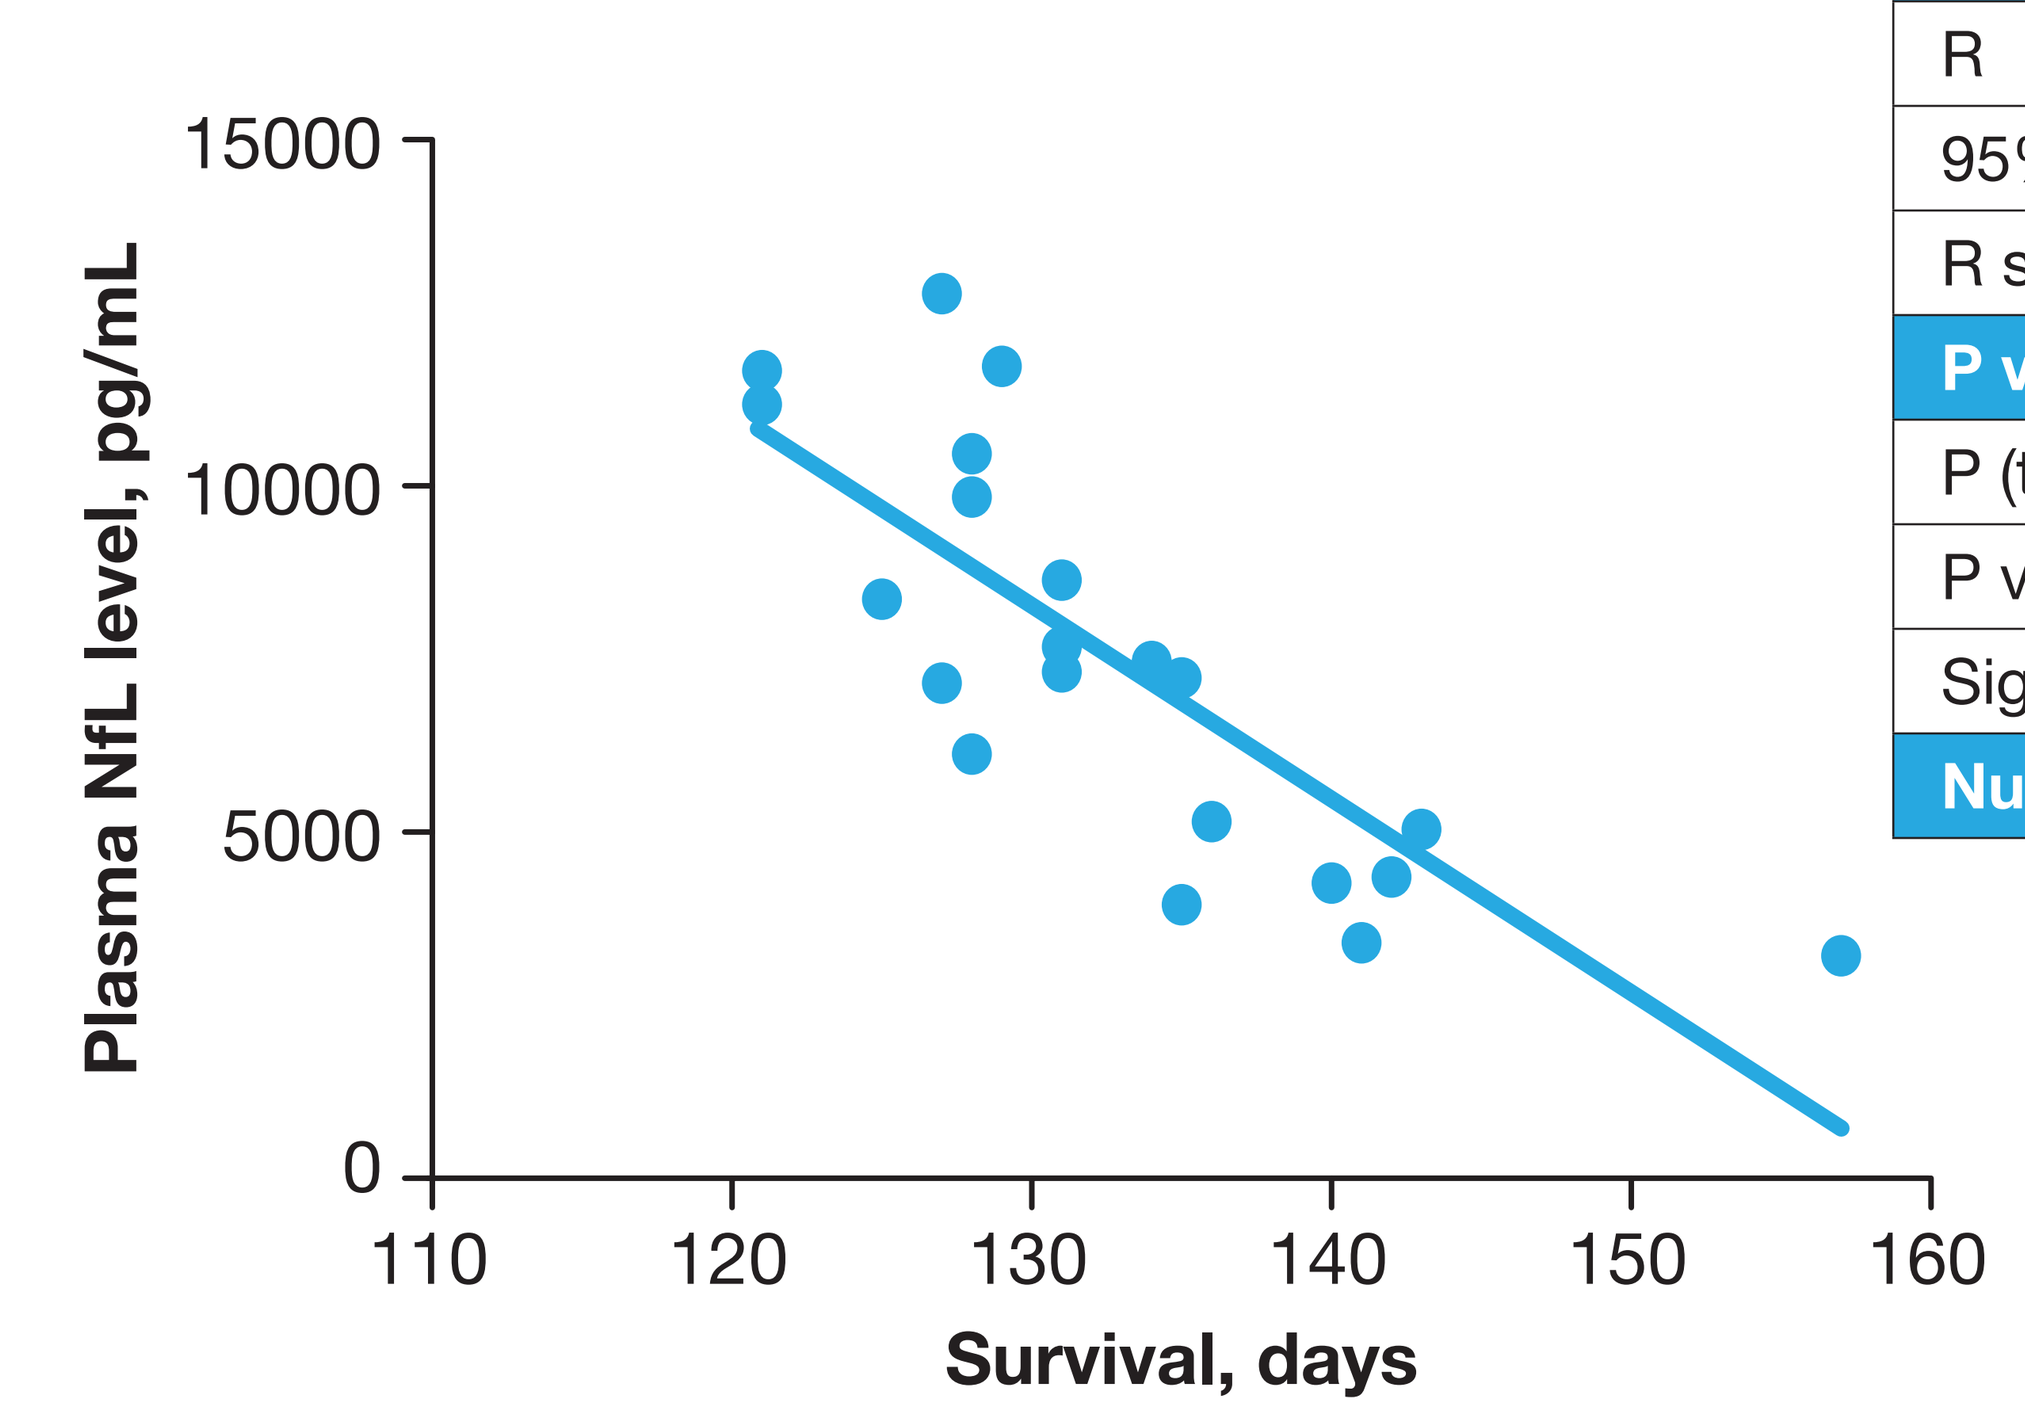
Plasma NfL level, pg/mL
Survival, days

Pearson r	
R	-0.7953
95% confidence interval	-0.9134 to -0.5538
R squared	0.6325
P value	
P (two-tailed)	<0.0001
P value summary	****
Significant? (alpha = 0.05)	Yes
Number of XY Pairs	**21**

CONCLUSIONS

- SBT-272-treated ALS mice experienced a dose dependent delay in the onset of neurological disease, a reduction in systemic markers of neurodegeneration and prolonged lifespan
 - Notably, these effects were sex dependent, despite no overt differences in drug exposure between sexes (data not shown)
 - The reason for this observed sex difference is currently unknown, although biomarker analyses corroborate a milder disease course in affected female mice which is consistent with survival data
- Pending histological analyses should shed further insight into the mechanism of SBT-272 neuroprotection in male ALS mice
- The data presented here establish SBT-272 as a potential therapeutic agent, with mitochondria as a tractable target for the treatment of patients with ALS
- Additional studies in TDP-43 model systems are currently planned to corroborate findings seen in the SOD1 model and to gain additional mechanistic insights

Study Funding: Trial funded by Stealth BioTherapeutics, Newton, MA

Acknowledgements: Medical writing assistance was provided by James A. Shiffer, RPh, Write On Time Medical Communications, LLC

To view this poster on the Stealth BioTherapeutics website, snap a photo of this QR code with your smart phone (QR reader required).

Stealth BIOTHERAPEUTICS